Association of the lipid biomarker, PCPro, and clinical outcomes in the ENZAMET trial (ANZUP 1304)

Lisa G Horvath[1,2,7.11.12], Hui-Ming Lin[2,7,11], Ian D Davis[3,7], Andrew J Martin[7,13], Tahlia Scheinberg[1,2,7,12], Neil Portman[2,7,11], Peter J Meikle[5], Kevin Huynh[5], Anthony Joshua[6,7,11], Margaret McJannett[7], Vinod Subhash[7], Sonia Yip[4,7], Scott A North[7,8], Raymond S McDermott[7,9], Kim N Chi[10], Martin R Stockler[4,7], Christopher Sweeney[7,14] for the ENZAMET trial investigators on behalf of the Australian and New Zealand Urogenital and Prostate Cancer Trials Group (ANZUP).

Affiliations: [1]Chris O'Brien Lifehouse, Australia; [2]Garvan Institute of Medical Research, Australia; [3]Monash University Eastern Health Clinical School, Australia; [4]NHMRC Clinical Trials Centre, University of Sydney, Australia; [5]Baker Heart and Diabetes Institute, Australia; [6]St Vincent's Hospital Sydney, Australia; [7]ANZUP, Australia; [8]University of Alberta, Canada; [9]St Vincent's University Hospital Dublin, Ireland; [10]BC Vancouver, Canada; [11]UNSW School of Clinical Medicine, Australia; [12]University of Sydney, Australia; [13]University of Queensland Centre for Clinical Research, Australia; [13]South Australian immunoGENomics Cancer Institute, Australia.

1. Background

Enzalutamide (ENZ)

- Improves overall survival (OS) in metastatic hormone sensitive prostate cancer (mHSPC).[1]
- Resistance is a problem – 11% of mHSPC patients on ENZ plus androgen deprivation therapy (ADT) die within 2 years of therapy.[1]

ENZAMET, a randomised Phase 3 trial

- Demonstrated benefit of ENZ plus ADT over conventional non-steroidal anti-androgen (NSAA) plus ADT in mHSPC.[1]

PCPro, a prognostic plasma lipid biomarker

- Validated for metastatic castration-resistant prostate cancer (mCRPC).[2]
- PCPro-positive = shorter OS & radiological progression-free survival.[2]
- Assay of 3 ceramides, cholesterol & triglycerides.[2]

PCPro-positive:

10.0506 × Cer(d18:1/18:0) + (-0.2783 × Cholesterol) + (-0.2240 × Triglycerides) + (-0.2979 × [Cer(d18:1/24:0)/Cer(d18:1/24:1)]) > *-1.1903*

2. Aim

To determine if PCPro is prognostic and/or predictive of enzalutamide response in mHSPC by post-hoc analysis of ENZAMET.

3. Methods

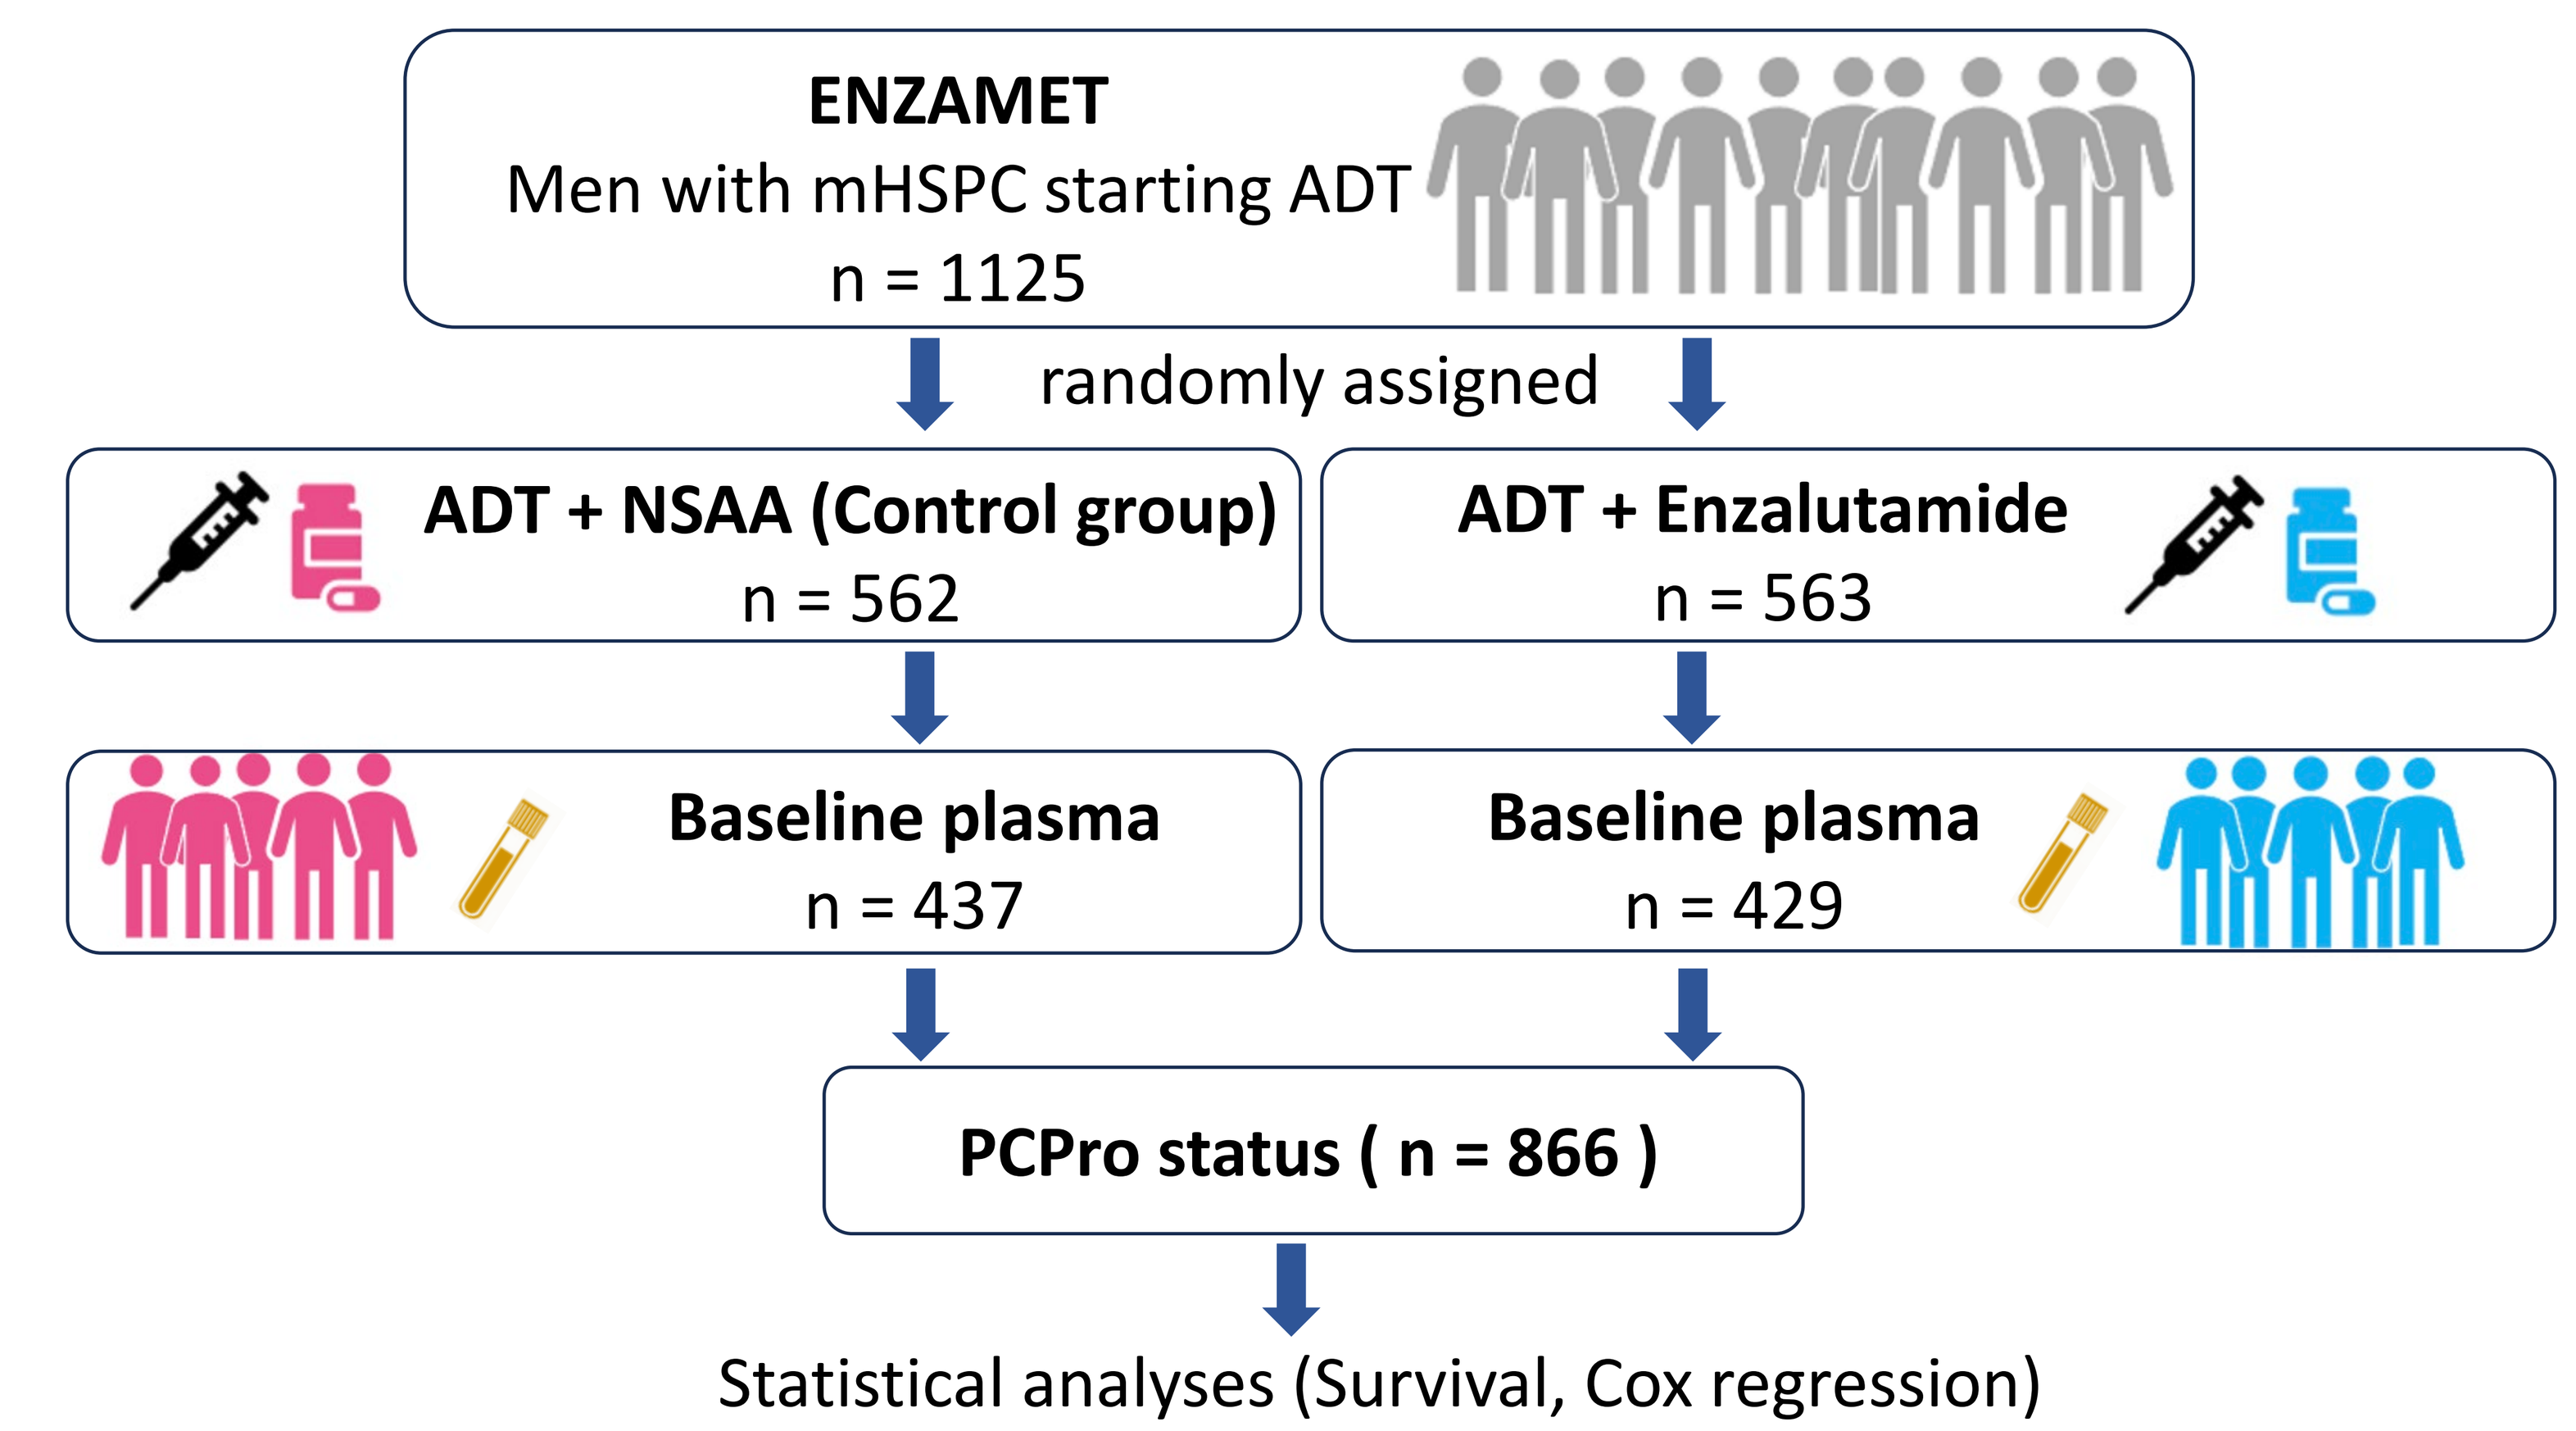
ENZAMET
Men with mHSPC starting ADT
n = 1125
randomly assigned
ADT + NSAA (Control group)
n = 562
ADT + Enzalutamide
n = 563
Baseline plasma
n = 437
Baseline plasma
n = 429
PCPro status (n = 866)
Statistical analyses (Survival, Cox regression)

4. Results

Table 1. Baseline clinical characteristics of study cohort by treatment arm.

Baseline characteristic	NSAA, N = 437 n (%)	ENZ, N = 429 n (%)	p-value[1]
PCPro			0.4
Negative	374 (86%)	376 (88%)	
Positive	63 (14%)	53 (12%)	
Volume of disease			>0.9
Low	203 (46%)	201 (47%)	
High	234 (54%)	228 (53%)	
Concurrent docetaxel	193 (44%)	194 (45%)	0.8
ACE-27 score			0.8
0-1	325 (74%)	323 (75%)	
2-3	112 (26%)	106 (25%)	
Anti-resorptive therapy	32 (7.3%)	32 (7.5%)	>0.9
Region			0.078
ANZ	277 (63%)	291 (68%)	
Ireland/UK	49 (11%)	56 (13%)	
N America	111 (25%)	82 (19%)	
Gleason grade			0.3
≤ 7	126 (34%)	112 (30%)	
8-10	249 (66%)	258 (70%)	
Unknown	62	59	
ECOG performance score			0.7
0	319 (73%)	309 (72%)	
1-2	118 (27%)	120 (28%)	
Age			0.3
<70	245 (56%)	226 (53%)	
≥ 70	192 (44%)	203 (47%)	
Visceral metastases	54 (12%)	49 (11%)	0.7
Metastatic status			0.2
M0 (metachronous)/ unknown	158 (36%)	172 (40%)	
M1 (synchronous)	279 (64%)	257 (60%)	

[1]Pearson's Chi-squared test

Table 2. Baseline clinical characteristics of study cohort by PCPro status.

Baseline characteristic	PCPro-negative, N = 750 n (%)	PCPro-positive, N = 116 n (%)	p-value[1]
Randomised to treatment arm			0.4
Conventional NSAA	374 (50%)	63 (54%)	
Enzalutamide	376 (50%)	53 (46%)	
Volume of disease			0.043
Low	360 (48%)	44 (38%)	
High	390 (52%)	72 (62%)	
Concurrent docetaxel	330 (44%)	57 (49%)	0.3
ACE-27 score			0.7
0-1	563 (75%)	85 (73%)	
2-3	187 (25%)	31 (27%)	
Anti-resorptive therapy	53 (7.1%)	11 (9.5%)	0.4
Region			0.2
ANZ	494 (66%)	74 (64%)	
Ireland/UK	85 (11%)	20 (17%)	
N America	171 (23%)	22 (19%)	
Gleason grade			0.7
≤ 7	210 (32%)	28 (30%)	
8-10	443 (68%)	64 (70%)	
Unknown	97	24	
ECOG performance score			0.042
0	553 (74%)	75 (65%)	
1-2	197 (26%)	41 (35%)	
Age			0.2
<70	414 (55%)	57 (49%)	
≥ 70	336 (45%)	59 (51%)	
Visceral metastases	82 (11%)	21 (18%)	0.026
Metastatic status			0.059
M0 (metachronous)/ unknown	295 (39%)	35 (30%)	
M1 (synchronous)	455 (61%)	81 (70%)	

[1]Pearson's Chi-squared test

Fig 1. PCPro-positive patients have shorter OS & clinical progression-free survival (clinPFS), indicating that PCPro is prognostic.

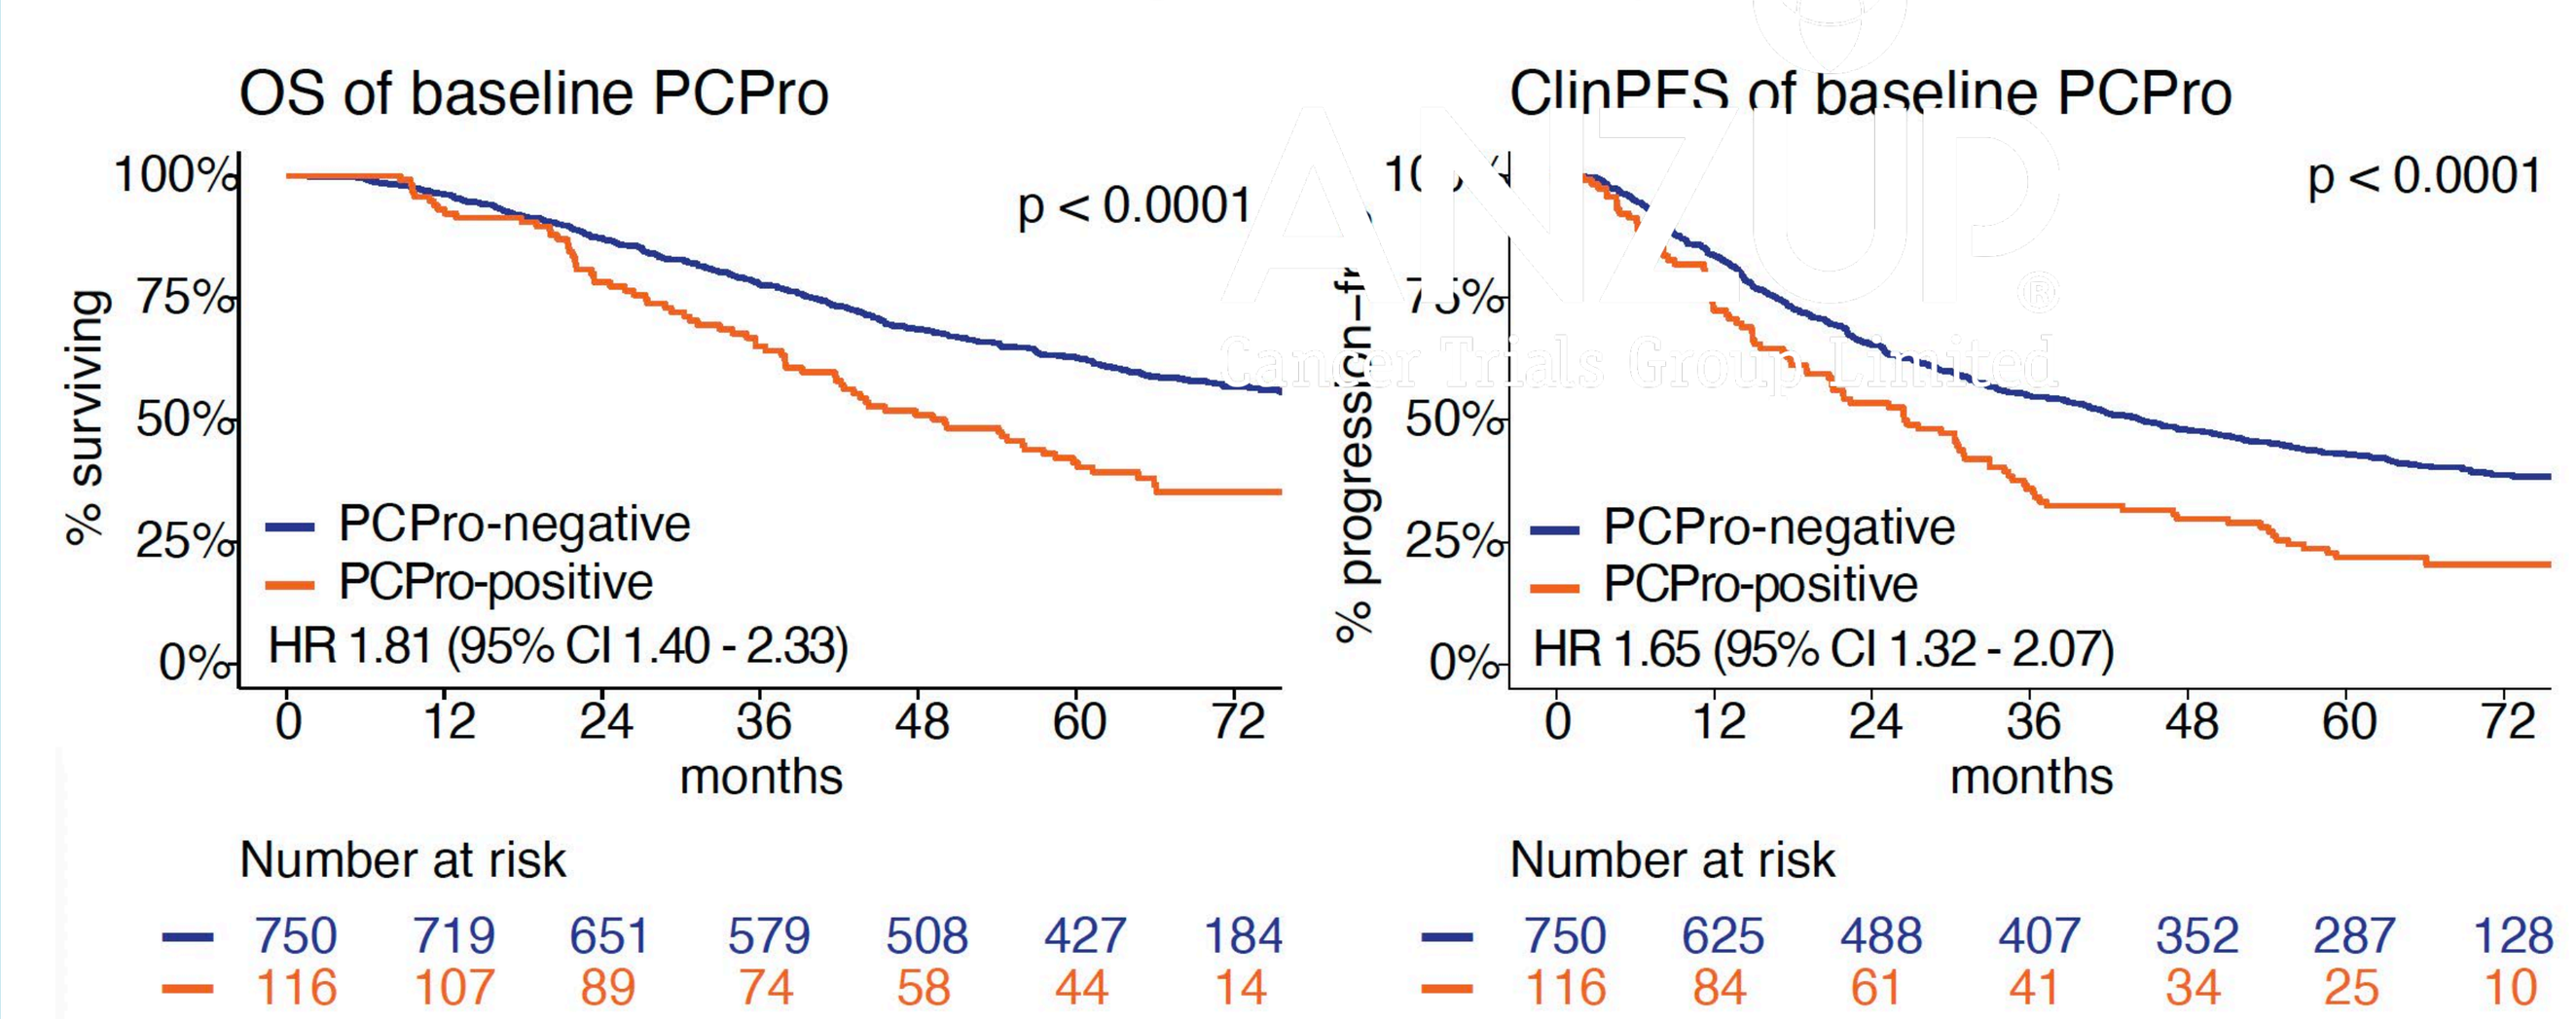
OS of baseline PCPro
p < 0.0001
% surviving
PCPro-negative
PCPro-positive
HR 1.81 (95% CI 1.40 - 2.33)
months
Number at risk
750 719 651 579 508 427 184
116 107 89 74 58 44 14
ClinPFS of baseline PCPro
p < 0.0001
PCPro-negative
PCPro-positive
HR 1.65 (95% CI 1.32 - 2.07)
months
Number at risk
750 625 488 407 352 287 128
116 84 61 41 34 25 10

Baseline	Median OS, month (95% CI)	5 year OS (95% CI)	Median ClinPFS, month (95% CI)	5 year ClinPFS (95% CI)
PCPro-negative	na	63% (59 - 66%)	44 (39 - 52)	43% (40 - 47%)
PCPro-positive	50 (42 - 61)	41% (33 - 51%)	26 (21 - 34)	22% (15 - 31%)

Fig 2. PCPro is an independent prognostic factor of OS & clinPFS when modeled with clinical variables in multivariable Cox regression analysis.

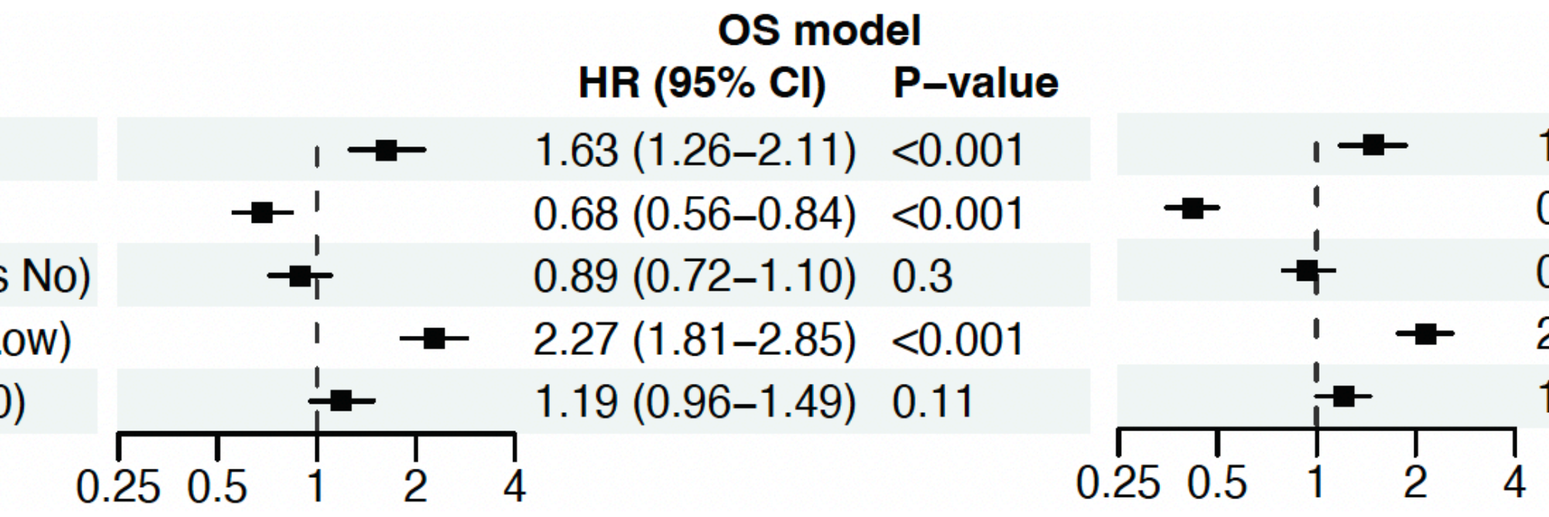

Variable in model	OS model HR (95% CI)	OS model P–value	ClinPFS model HR (95% CI)	ClinPFS model P–value
Baseline PCPro ([+] vs [-])	1.63 (1.26–2.11)	<0.001	1.49 (1.18–1.86)	<0.001
Treatment (Enza vs NSAA)	0.68 (0.56–0.84)	<0.001	0.42 (0.35–0.50)	<0.001
Concurrent docetaxel (Yes vs No)	0.89 (0.72–1.10)	0.3	0.94 (0.79–1.13)	0.5
Volume of disease (High vs Low)	2.27 (1.81–2.85)	<0.001	2.14 (1.77–2.58)	<0.001
Stage at diagnosis (M1 vs M0)	1.19 (0.96–1.49)	0.11	1.21 (1.00–1.45)	0.045

Fig 3. There is a significant interaction between PCPro and treatment in Cox regression of OS but not for clinPFS, indicating that the OS benefit of ENZ differs by PCPro status.

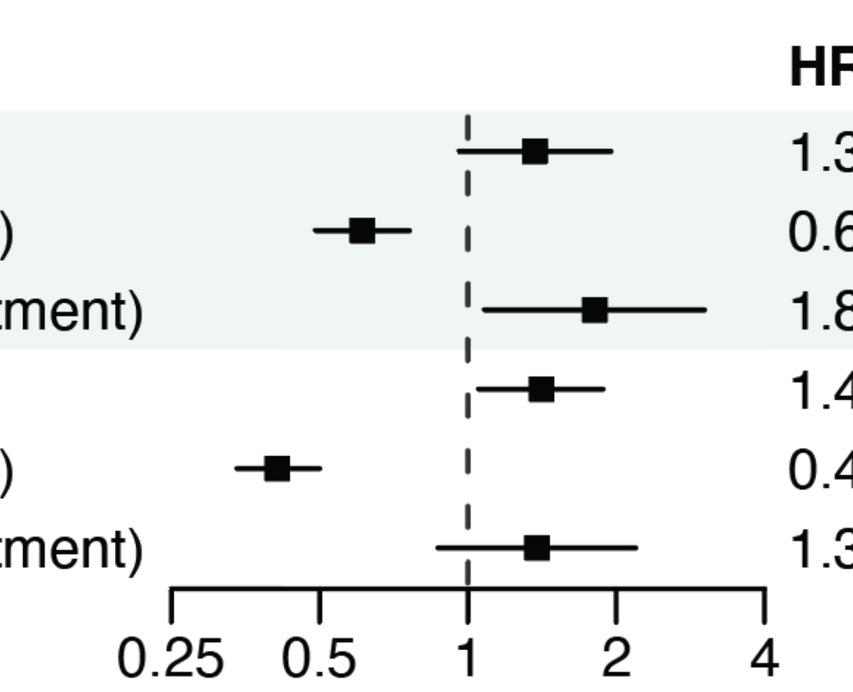

Model	Variable	HR (95% CI)	P–value
OS	Baseline PCPro ([+] vs [-])	1.37 (0.96–1.95)	0.082
	Treatment (Enza vs NSAA)	0.61 (0.49–0.76)	<0.001
	Interaction (PCPro × Treatment)	1.81 (1.08–3.02)	0.024
ClinPFS	Baseline PCPro ([+] vs [-])	1.41 (1.05–1.88)	0.021
	Treatment (Enza vs NSAA)	0.41 (0.34–0.50)	<0.001
	Interaction (PCPro × Treatment)	1.38 (0.87–2.19)	0.2

Fig 4. OS of PCPro-positive patients was not improved by ENZ when compared to NSAA, unlike PCPro-negative patients, indicating that PCPro-positive patients are resistant to ENZ.

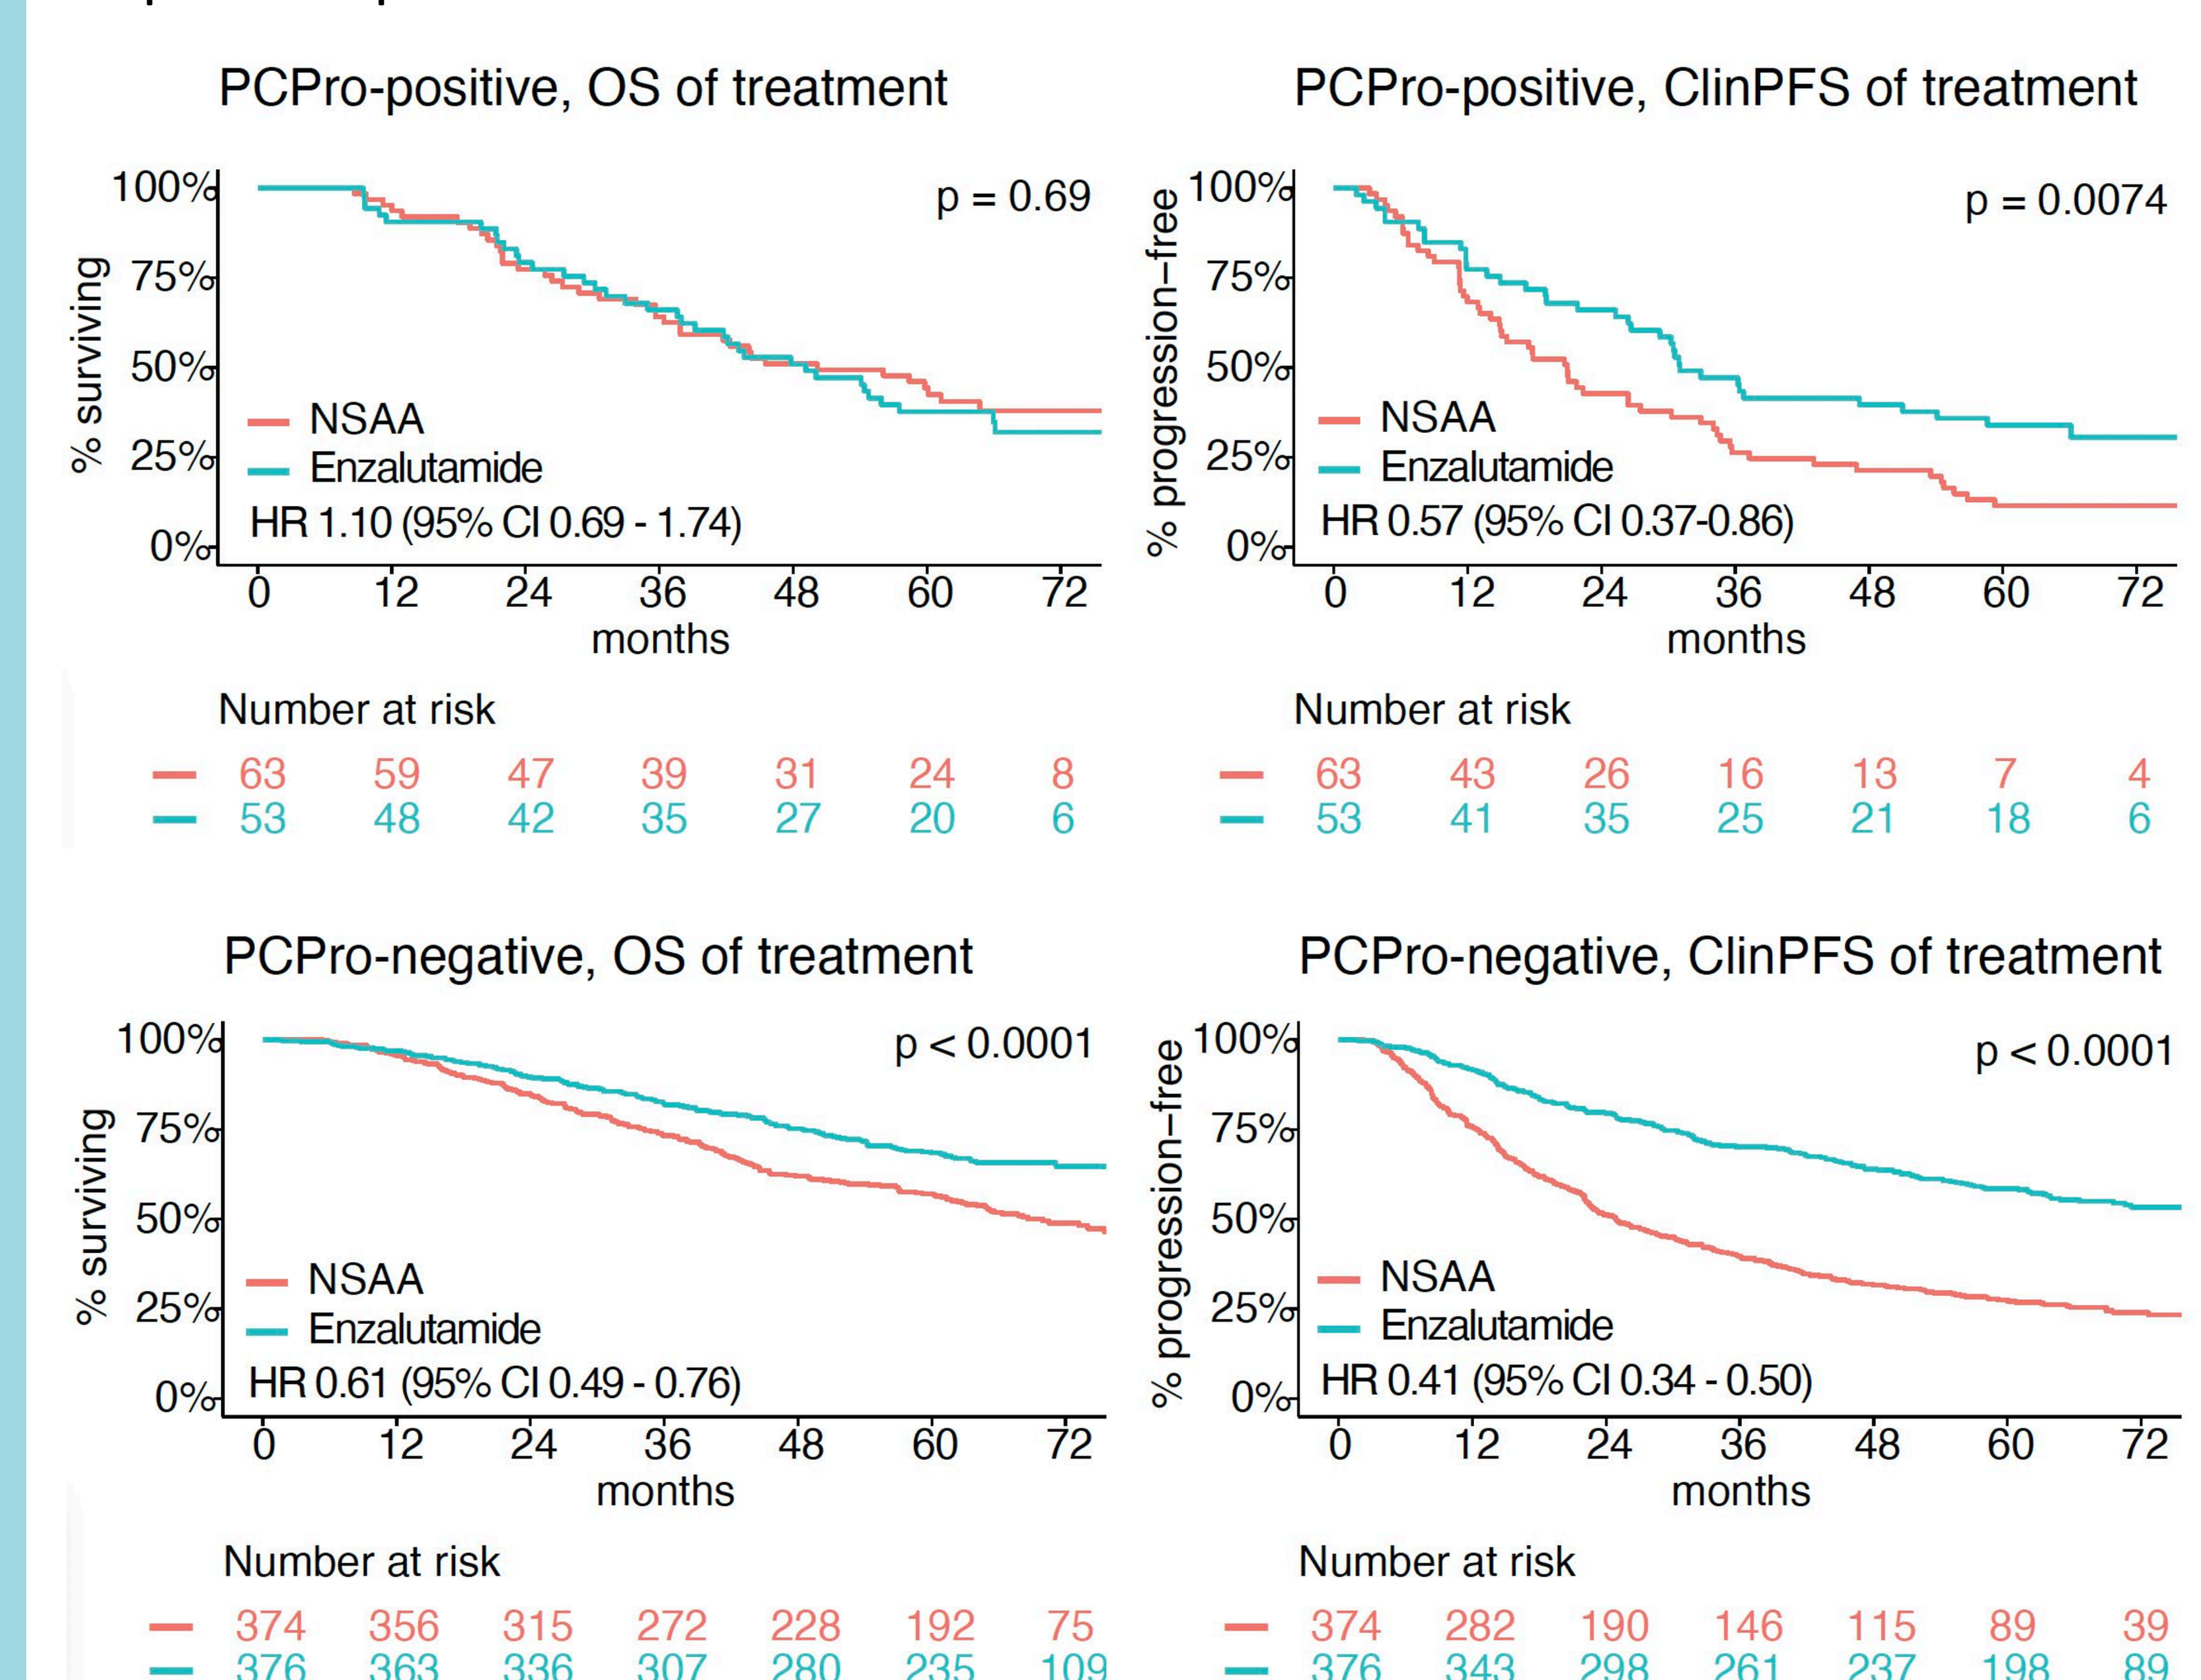
PCPro-positive, OS of treatment
p = 0.69
NSAA
Enzalutamide
HR 1.10 (95% CI 0.69 - 1.74)
Number at risk
63 59 47 39 31 24 8
53 48 42 35 27 20 6
PCPro-positive, ClinPFS of treatment
p = 0.0074
HR 0.57 (95% CI 0.37-0.86)
63 43 26 16 13 7 4
53 41 35 25 21 18 6
PCPro-negative, OS of treatment
p < 0.0001
HR 0.61 (95% CI 0.49 - 0.76)
374 356 315 272 228 192 75
376 363 336 307 280 235 109
PCPro-negative, ClinPFS of treatment
p < 0.0001
HR 0.41 (95% CI 0.34 - 0.50)
374 282 190 146 115 89 39
376 343 298 261 237 198 89

Baseline	Treatment	Median OS, month (95% CI)	5 year OS (95% CI)	Median ClinPFS, month (95% CI)	5 year ClinPFS (95% CI)
PCPro-positive	NSAA	50 (38 - na)	44% (33 - 59%)	21 (15 - 33)	12% (5.7 - 23%)
PCPro-positive	Enzalutamide	49 (39 - 66)	38% (27 - 53%)	31 (27 - 59)	34% (23 - 49%)
PCPro-negative	NSAA	70 (62 - na)	57% (52 - 62%)	25 (22 - 30)	27% (23 - 32%)
PCPro-negative	Enzalutamide	na	68% (64 - 73%)	81 (69 - na)	59% (54 - 64%)

5. Conclusions

PCPro-positive patients have shorter survival than PCPro-negative patients.

Despite ENZA improving clinPFS, PCPro-positive patients treated with ENZA have no OS benefit compared to those treated with NSAA.

6. Contact Us

lisa.horvath@lh.org.au & anzup@anzup.org.au

www.anzup.org.au

@ANZUPtrials

#ENZAMET

Acknowledgements

We thank our funders (NHMRC, Cancer Australia, Astellas Pharma Inc., Noel Castan Fellowship), study staff and trial participants.

References.

1. *Sweeney et al (2023) Lancet Oncol 24:323;*
2. *Scheinberg et al (2024) Prostate Cancer Prostatic Dis 27:136*

Abstract # 1607P